#340/15 The Hu2G10 tumor-selective anti–Trop-2 monoclonal antibody targets the cleaved-activated Trop-2 ablates growth of multiple human cancers

Saverio Alberti, MD, PhD[1,2*]; Marco Trerotola, PhD[3]; Emanuela Guerra, PhD[2,3]

www.mediterranea-theranostic.com/en/

[1]Unit of Medical Genetics, Department of Biomedical Sciences - BIOMORF, University of Messina, Messina, Italy [2]Mediterranea Theranostic, Lanciano, Italy [3]Laboratory of Cancer Pathology, CAST, and Department of Medical, Oral and Biotechnological Sciences, "G. d'Annunzio University" of Chieti-Pescara, Chieti, Italy *alberti.saverio@gmail.com

ABSTRACT

Trop-2 is a transmembrane glycoprotein that transduces calcium, PI and PKC signals (4) and activates growth-signaling networks that converge on Akt, ERK, Cyclin D1, NFkB (1). We discovered that Trop-2 cleavage by ADAM10 is an activator switch for induction of cancer growth and metastatic diffusion (2, 3). This cleavage causes a spatial rearrangement of the extracellular portion of Trop-2 and exposes protein sites that are normally inaccessible. We devised a strategy to obtain monoclonal antibodies (mAbs) capable of recognizing these cancer-exposed sites.

Most Trop-2-targeted approaches are based on mAbs that recognize a single immunodominant epitope, poised between the globular and stem regions. Such mAbs have limited therapeutic index.

We removed the immunodominant epitope by deletion mutagenesis. To optimize breadth of immune response, mice were immunized with target proteins produced in cells with multy-phyla end-glycosylation patterns, including tumor cells, insect, yeast, bacterial cells.

Cancer-specific mAbs were screened by cell-based ELISA assays, bio-layer interferometry, flow cytometry and fluorescence microscopy on cleaved, wild-type and glycosylation-site Trop-2 mutants. mAbs were obtained that efficiently bound Trop-2 expressing cancer cells and inhibited cell growth in vitro.

We humanized the 2G10 mAb (Hu2G10 IgG1/k) for an affinity for recombinant, cancer-specific, cleaved Trop-2 of $<10^{-12}$ M, i.e. 10,000-fold higher than for the wtTrop-2, and 1,000-fold higher than Trodelvy.

In vivo the naked Hu2G10 antibody ablated the growth of breast, colon, ovary and prostate cancers. In the ADC setting Hu2G10 drove high anticancer effectiveness with unprecedented specificity, for best-in-class anticancer therapy.

INTRODUCTION

Trop-2 activates pro-growth and metastasis signaling networks in cancer, where it is widely overexpressed and associated to poor prognosis (1,3,4).

Most anti-Trop-2 antibodies, among them RS7, Sacituzumab Govitecan (SG) ADC (TRODELVY, IMMU-132), bind the same, immunodominant epitope, located in the D146-L274 N-terminal region. SG shows some efficacy in TNBC patients, Corresponding promise was shown in patients with additional types of Trop-2-expressing cancers. However, the SG half-life is 11–14 hours in plasma and SG induces limiting side effects such as neutropenia and diarrhea, that are associated to ineffective retention of its SN38 payload in the circulation. The Rinat-Pfizer PF-06664178 anti-Trop-2 ADC was also shown to bind to the immunodominant epitope of Trop-2. Despite early promise PF-06664178 was ultimately discontinued because of excess toxicity with modest antitumor activity. Hence a major stumbling block for next-generation therapeutic strategies remains the vast expression of Trop-2 in normal tissues.

We found that activation of Trop-2 for driving cancer progression requires proteolytic activation by ADAM10. **This does not to occur in normal tissues** (2,3). We succeded in developing the Hu2G10 Mab that specifically targets the cancer-activated, proteolitycally-cleaved form of Trop-2.

References

1. Guerra et al. Oncogene 32, 1594-1600 (2013). 2. Trerotola et al. Neoplasia 23, 415-428 (2021). 3. Guerra et al. Neoplasia 23, 898-911 (2021). 4. Guerra et al. Oncogene 41, 1795-1808 (2022).

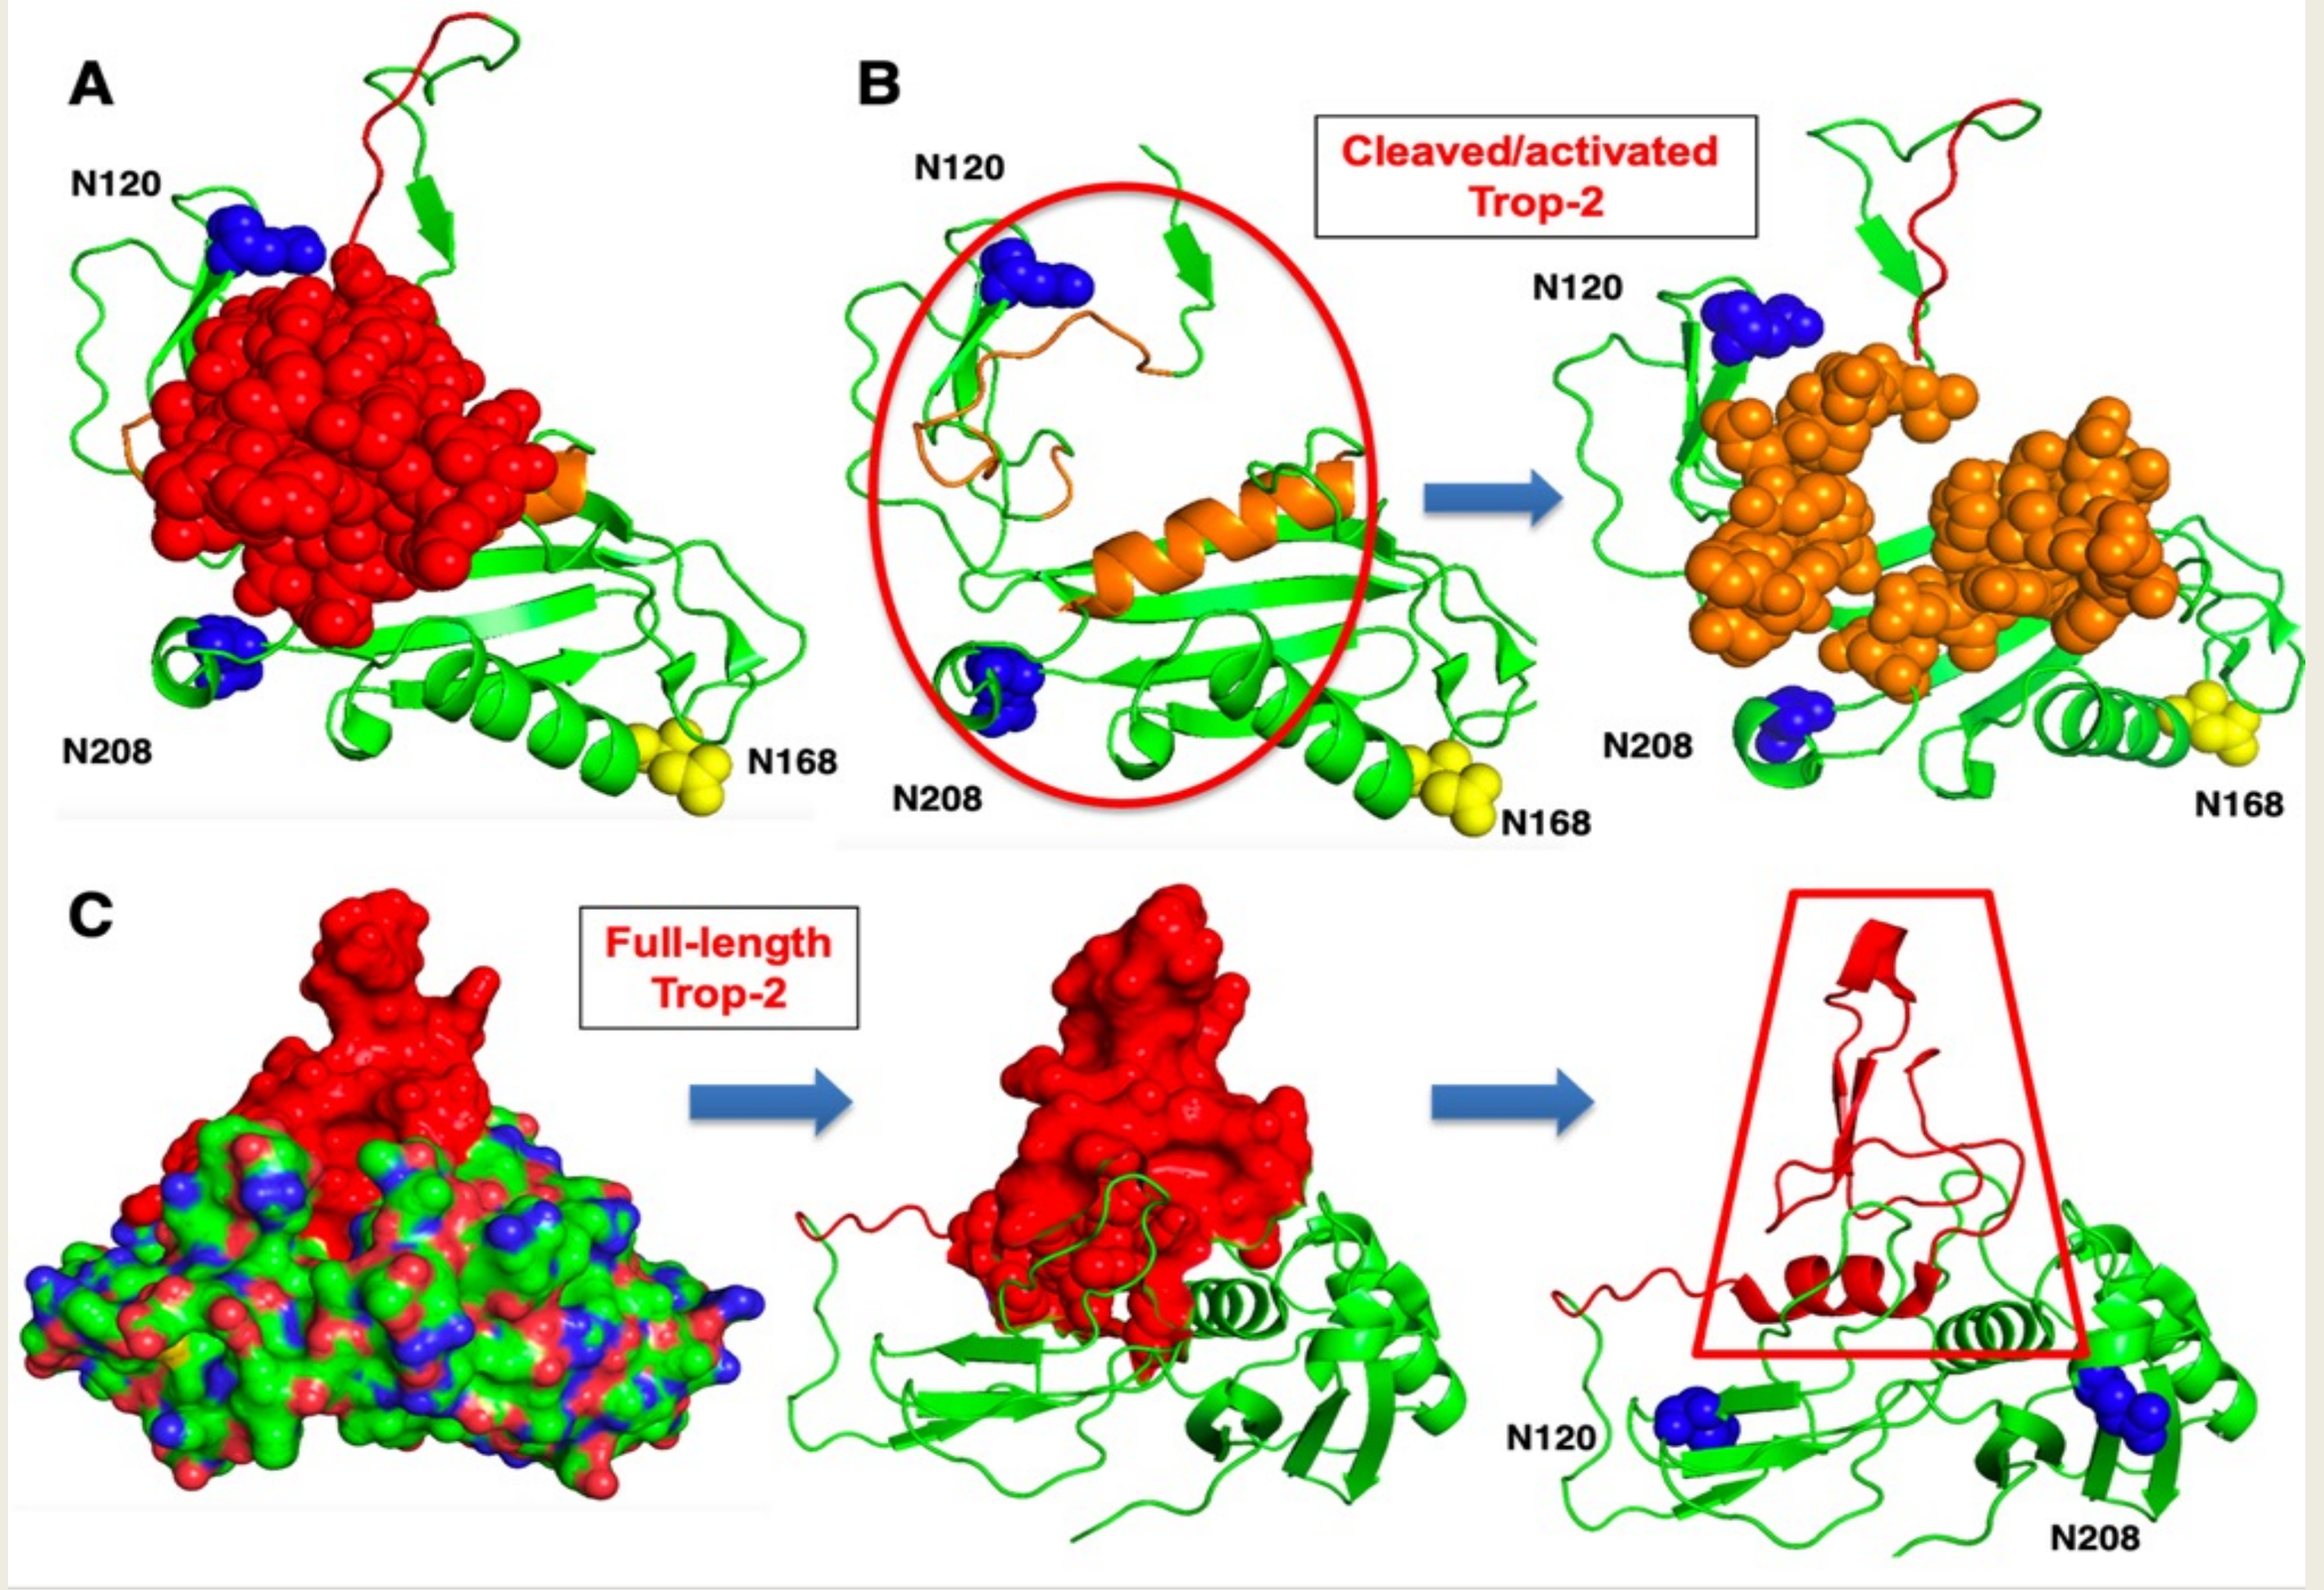
A
N120
N208
N168
B
Cleaved/activated Trop-2
C
Full-length Trop-2

Figure 1. 3D structure of Trop-2. (A) 3D structure of the extra-cellular domain of Trop-2 (Pavšič, 2021). The N-terminal subunit is in red, Trop-2 N-glycosylation sites are indicated. (B) 3D structure of the extra-cellular domain of Trop-2 devoid of the N-terminal subunit. (*left*) Red oval: region of the Trop-2 main chain contacted by the N-terminal subunit. (C) Full-length extra-cellular domain of Trop-2.

RESULTS

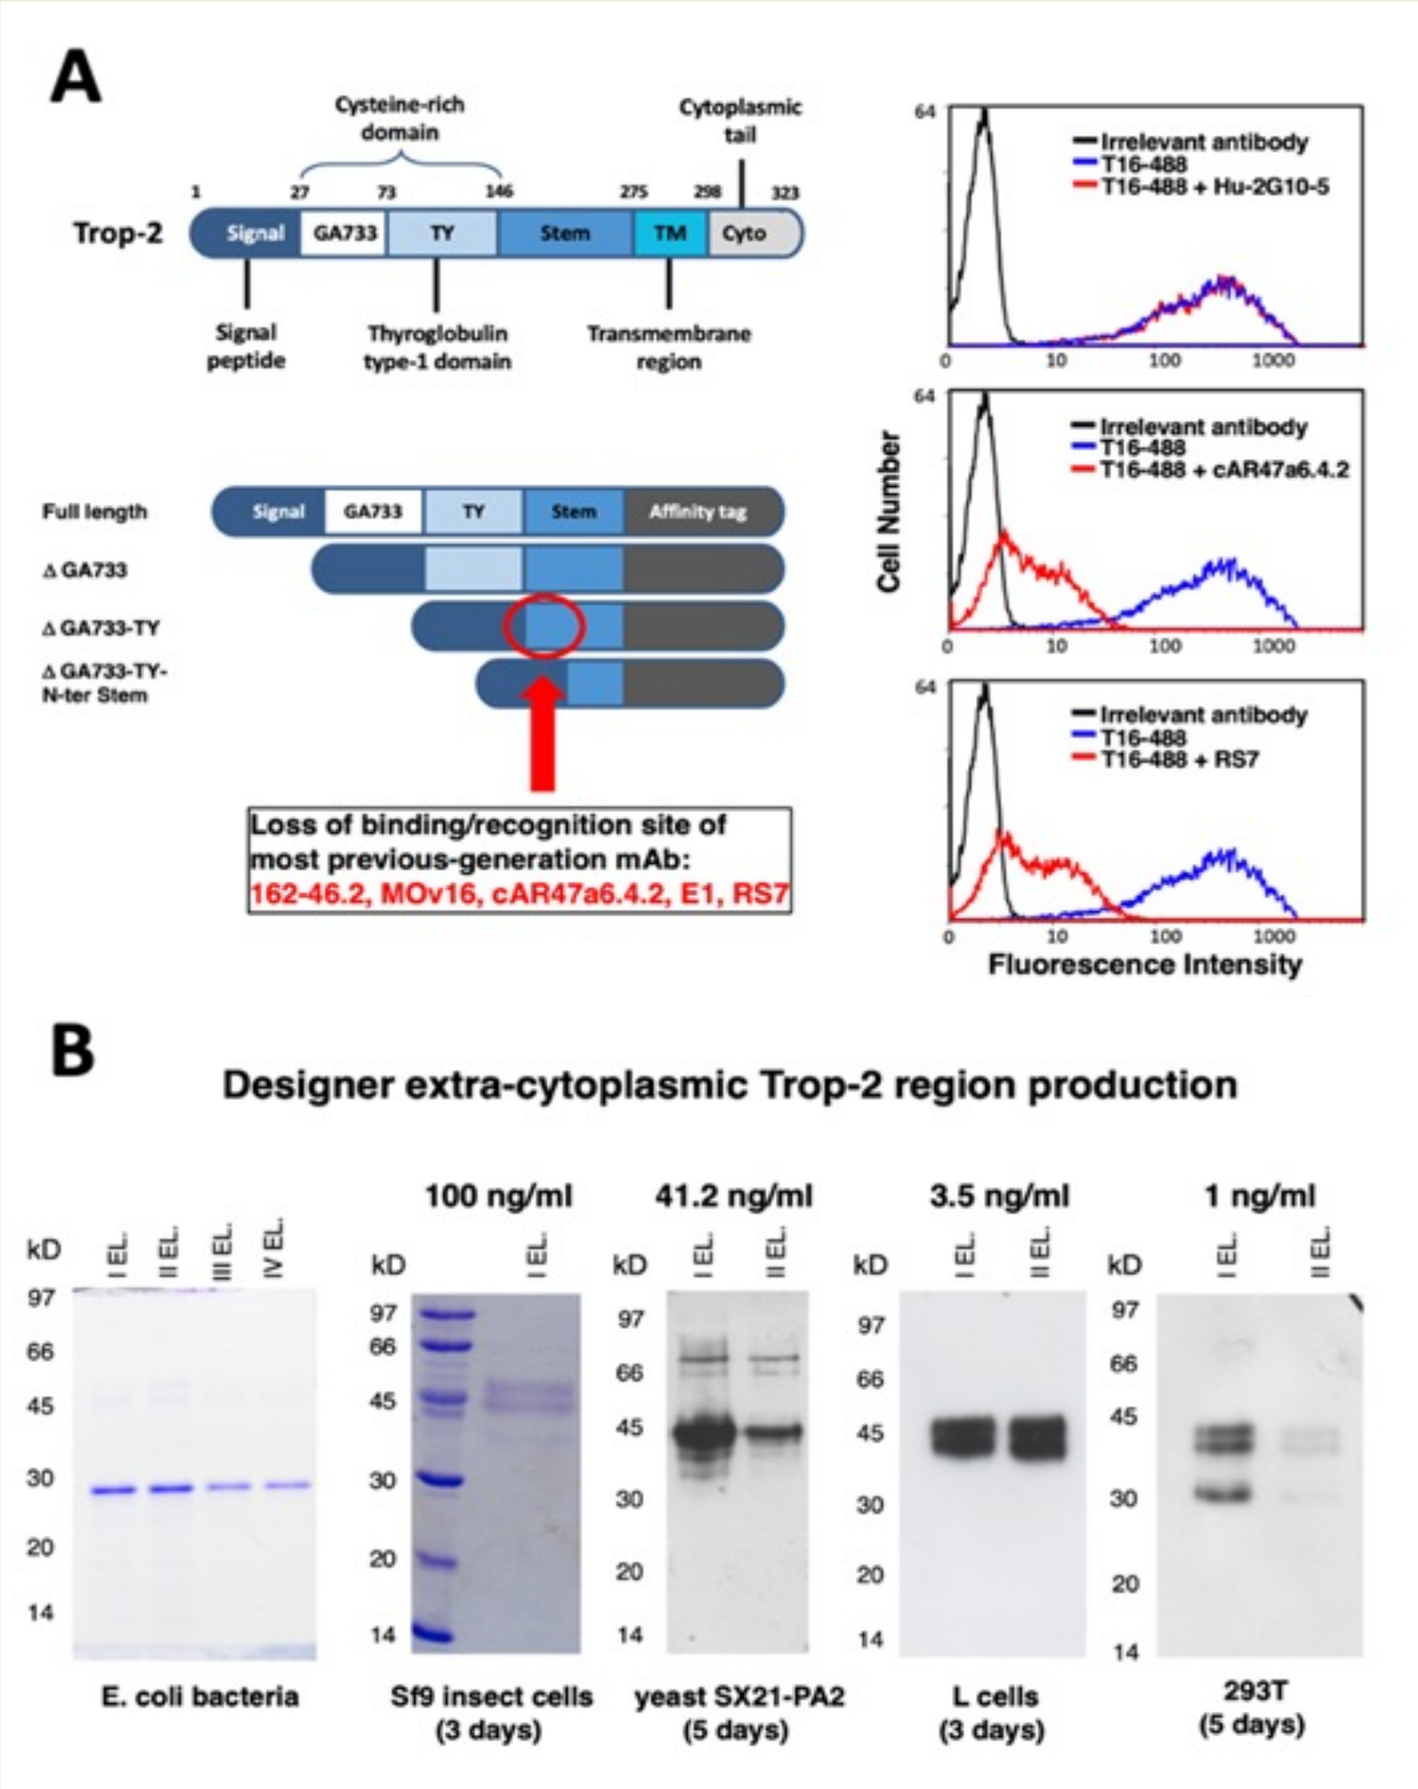
Loss of binding/recognition site of most previous-generation mAb: 162-46.2, MOv16, cAR47a6.4.2, E1, RS7
Fluorescence Intensity
Designer extra-cytoplasmic Trop-2 region production
E. coli bacteria
Sf9 insect cells (3 days)
yeast SX21-PA2 (5 days)
L cells (3 days)
293T (5 days)

Figure 2. Generation of mAb targeting cancer-activated Trop-2. (A) Immunodominant recognition of Trop-2 (modified from Ikeda, 2015). (left) Trop-2 schematics. (right) Flow cytometry analysis of competition assays versus immunodominant-epitope-binding mAbs. (B) Trop-2 recombinant protein immunogens

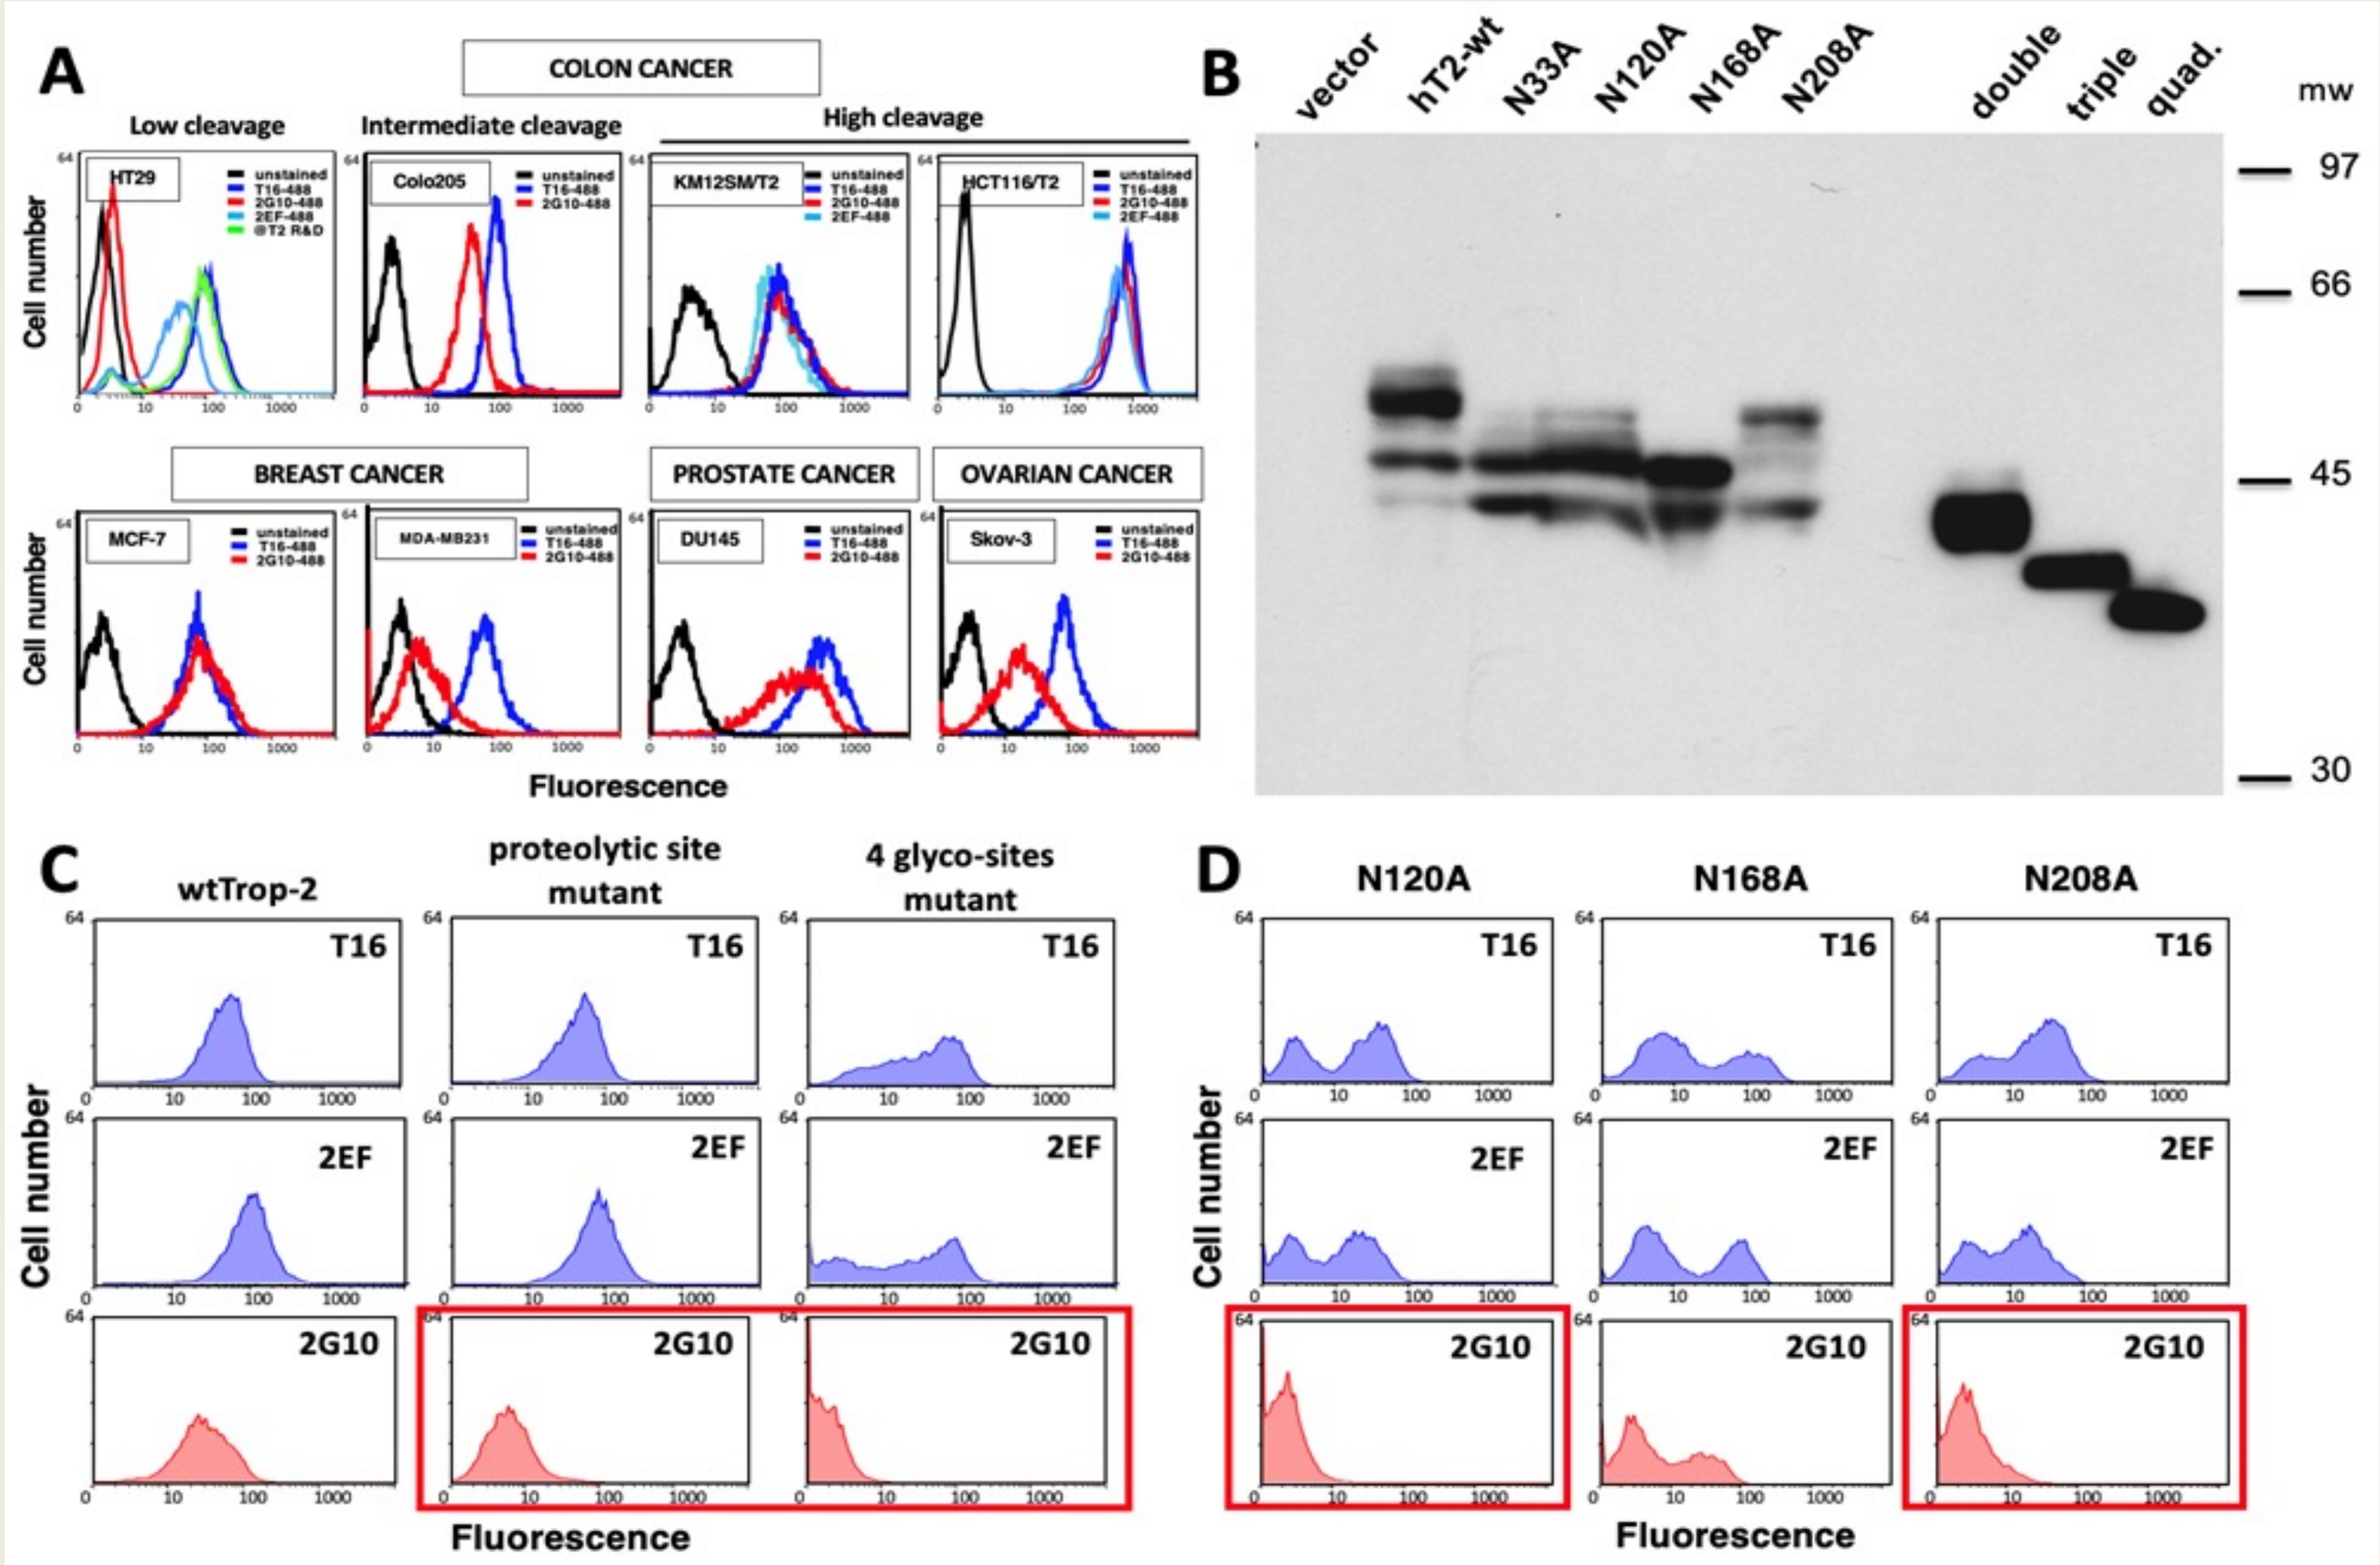
COLON CANCER
BREAST CANCER
PROSTATE CANCER
OVARIAN CANCER
wtTrop-2
proteolytic site mutant
4 glyco-sites mutant
N120A
N168A
N208A
Fluorescence

Figure 3. Glycosylation-dependent binding of 2G10 to Trop-2. (A) Flow cytometry analysis of 2G10 binding to wtTrop-2 in cancer cell lines with different extent of Trop-2 cleavage. (B) Western blot analysis of Trop-2 glycosylation mutants; wtTrop-2: full-glycosylation control. (C,D) Flow cytometry analysis of mutant-Trop-2 KM12SM transfectants for 2G10 binding. Benchmark T16, 2EF anti-framework Trop-2 mAb and R&D anti-Trop-2 pAb were utilized to quantify Trop-2 molecules at the cell membrane.

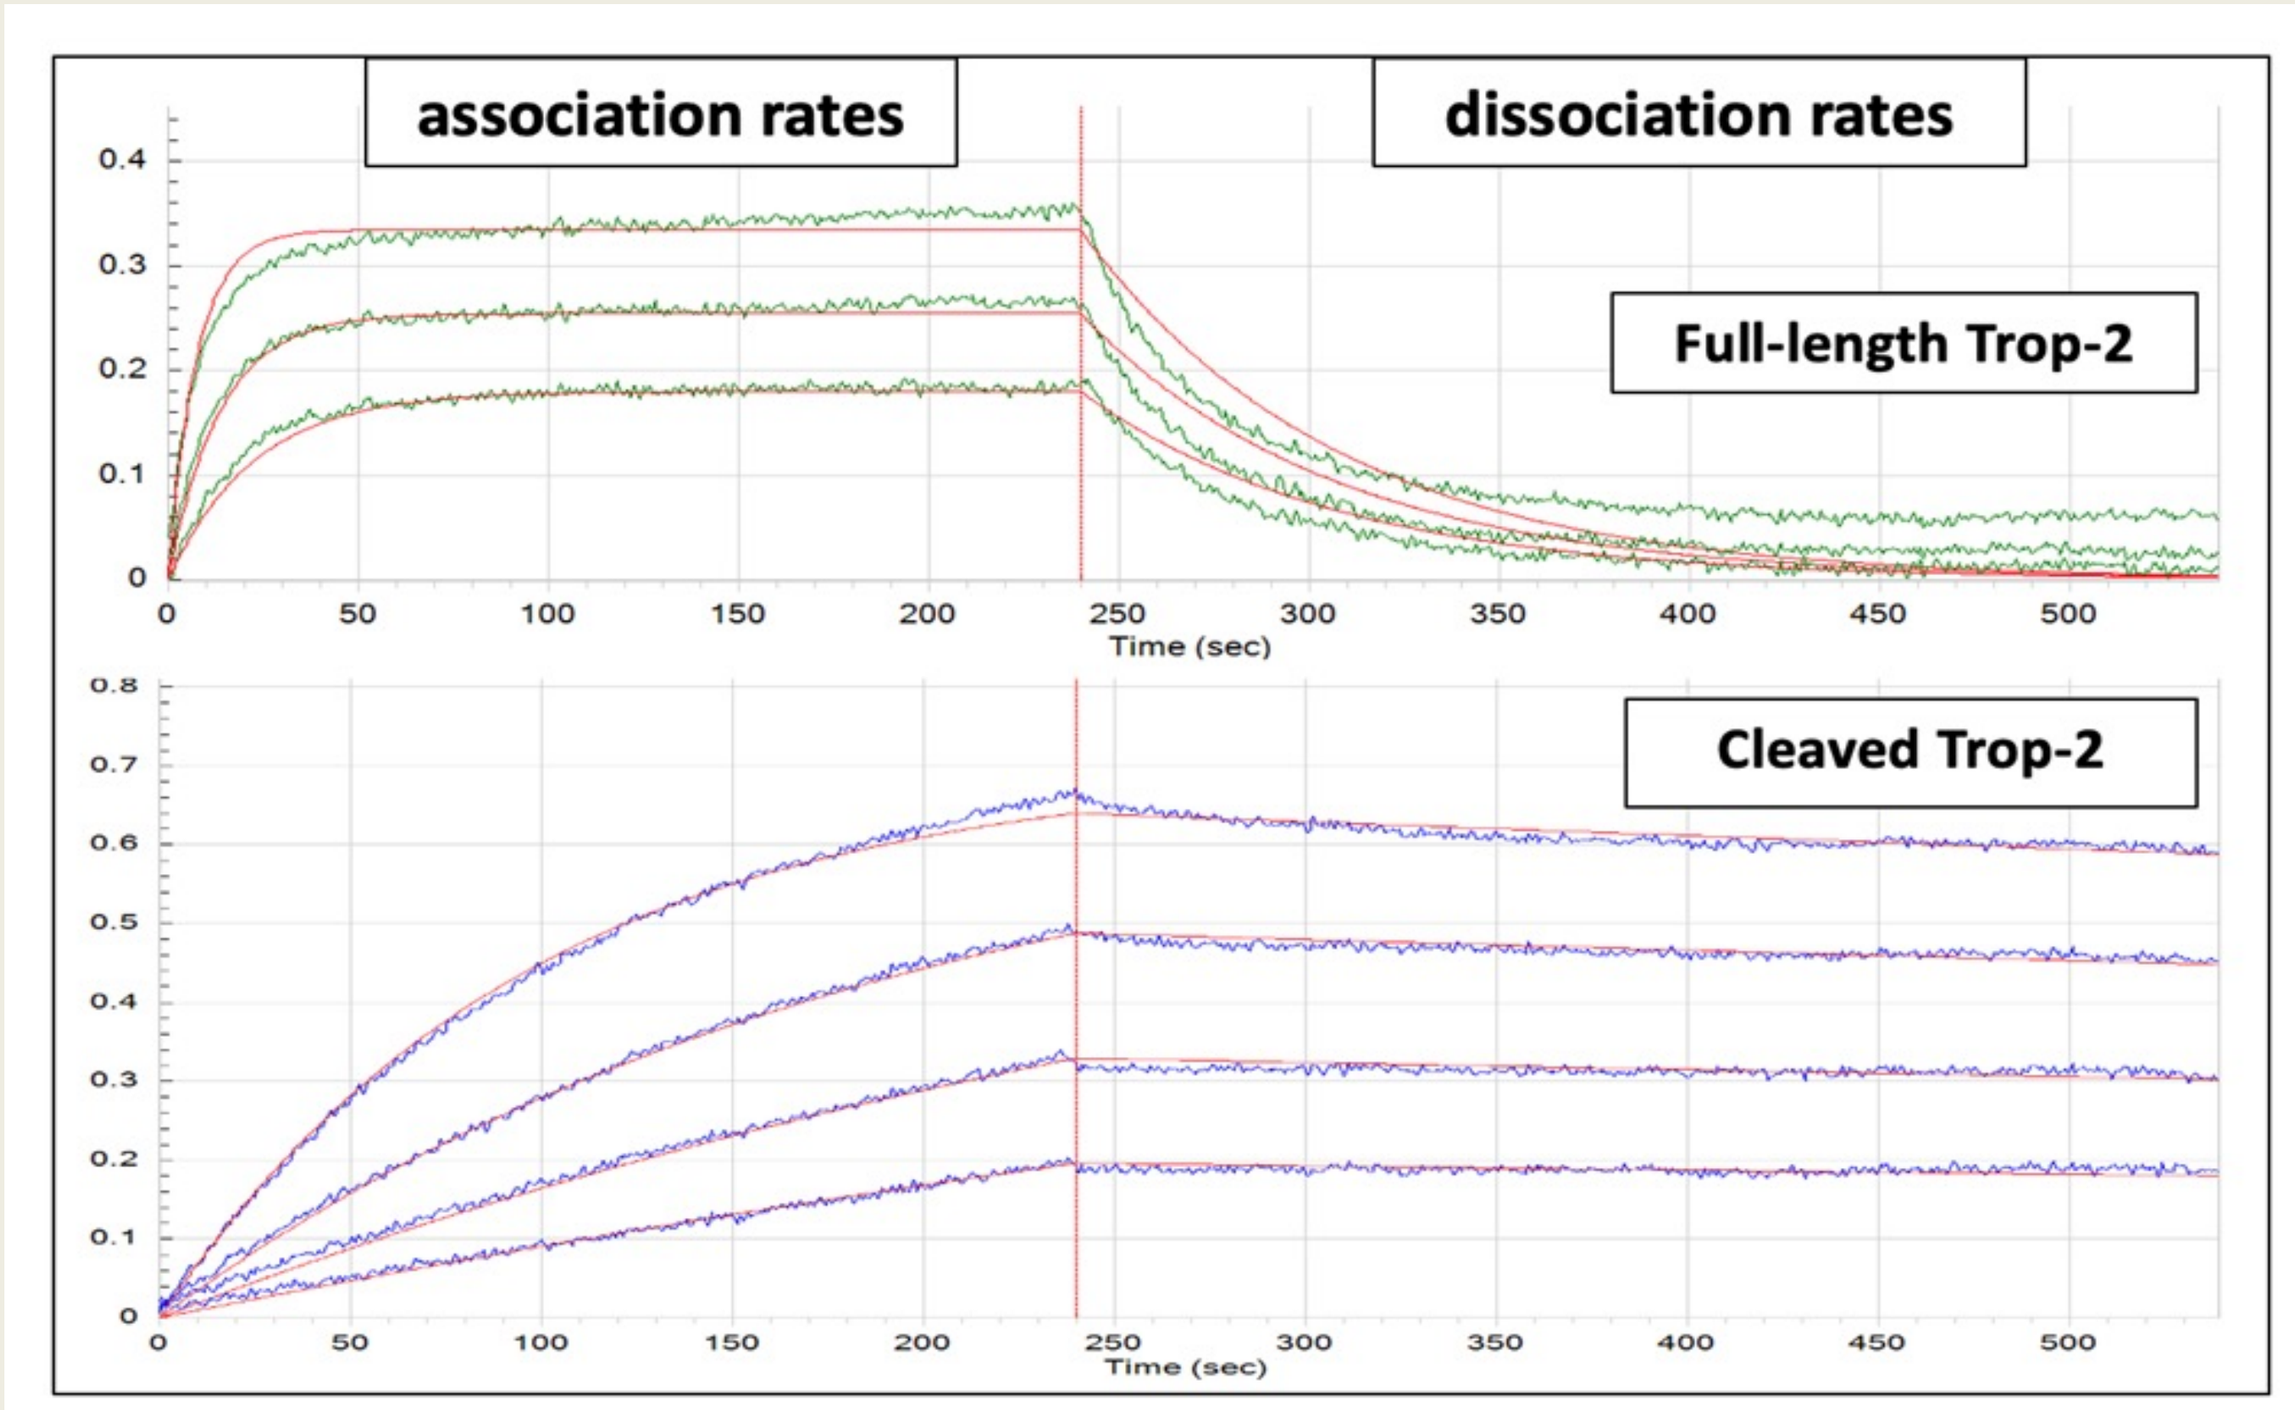
association rates
dissociation rates
Full-length Trop-2
Cleaved Trop-2
Time (sec)

Ligand	Analyte	Conc. (nM)	KD (M)	k on (1/Ms)	k dis (1/s)	χ^2	R^2
Hu 2G10	T2 –full length	250-62.5	3.16E-08	4.68E+05	1.48E-02	1.2872	0.9682
Hu 2G10	T2-Cleaved	25-6.25	<1.0E-12	1.90E+05	<1.0E-07	1.4568	0.9772

Figure 4. Hu2G10 affinity measurements. Hu2G10 mAb absolute affinity was measured by Bio-Layer Interferometry-based label-free binding. Association/dissociation kinetics are shown for the proteolysis resistant (top) and the fully cleaved (mid) Trop-2 ligands, plotted as wavelength shifts towards time with (association) or without (dissociation) analytes. Numerical values are shown from data fitting routines.

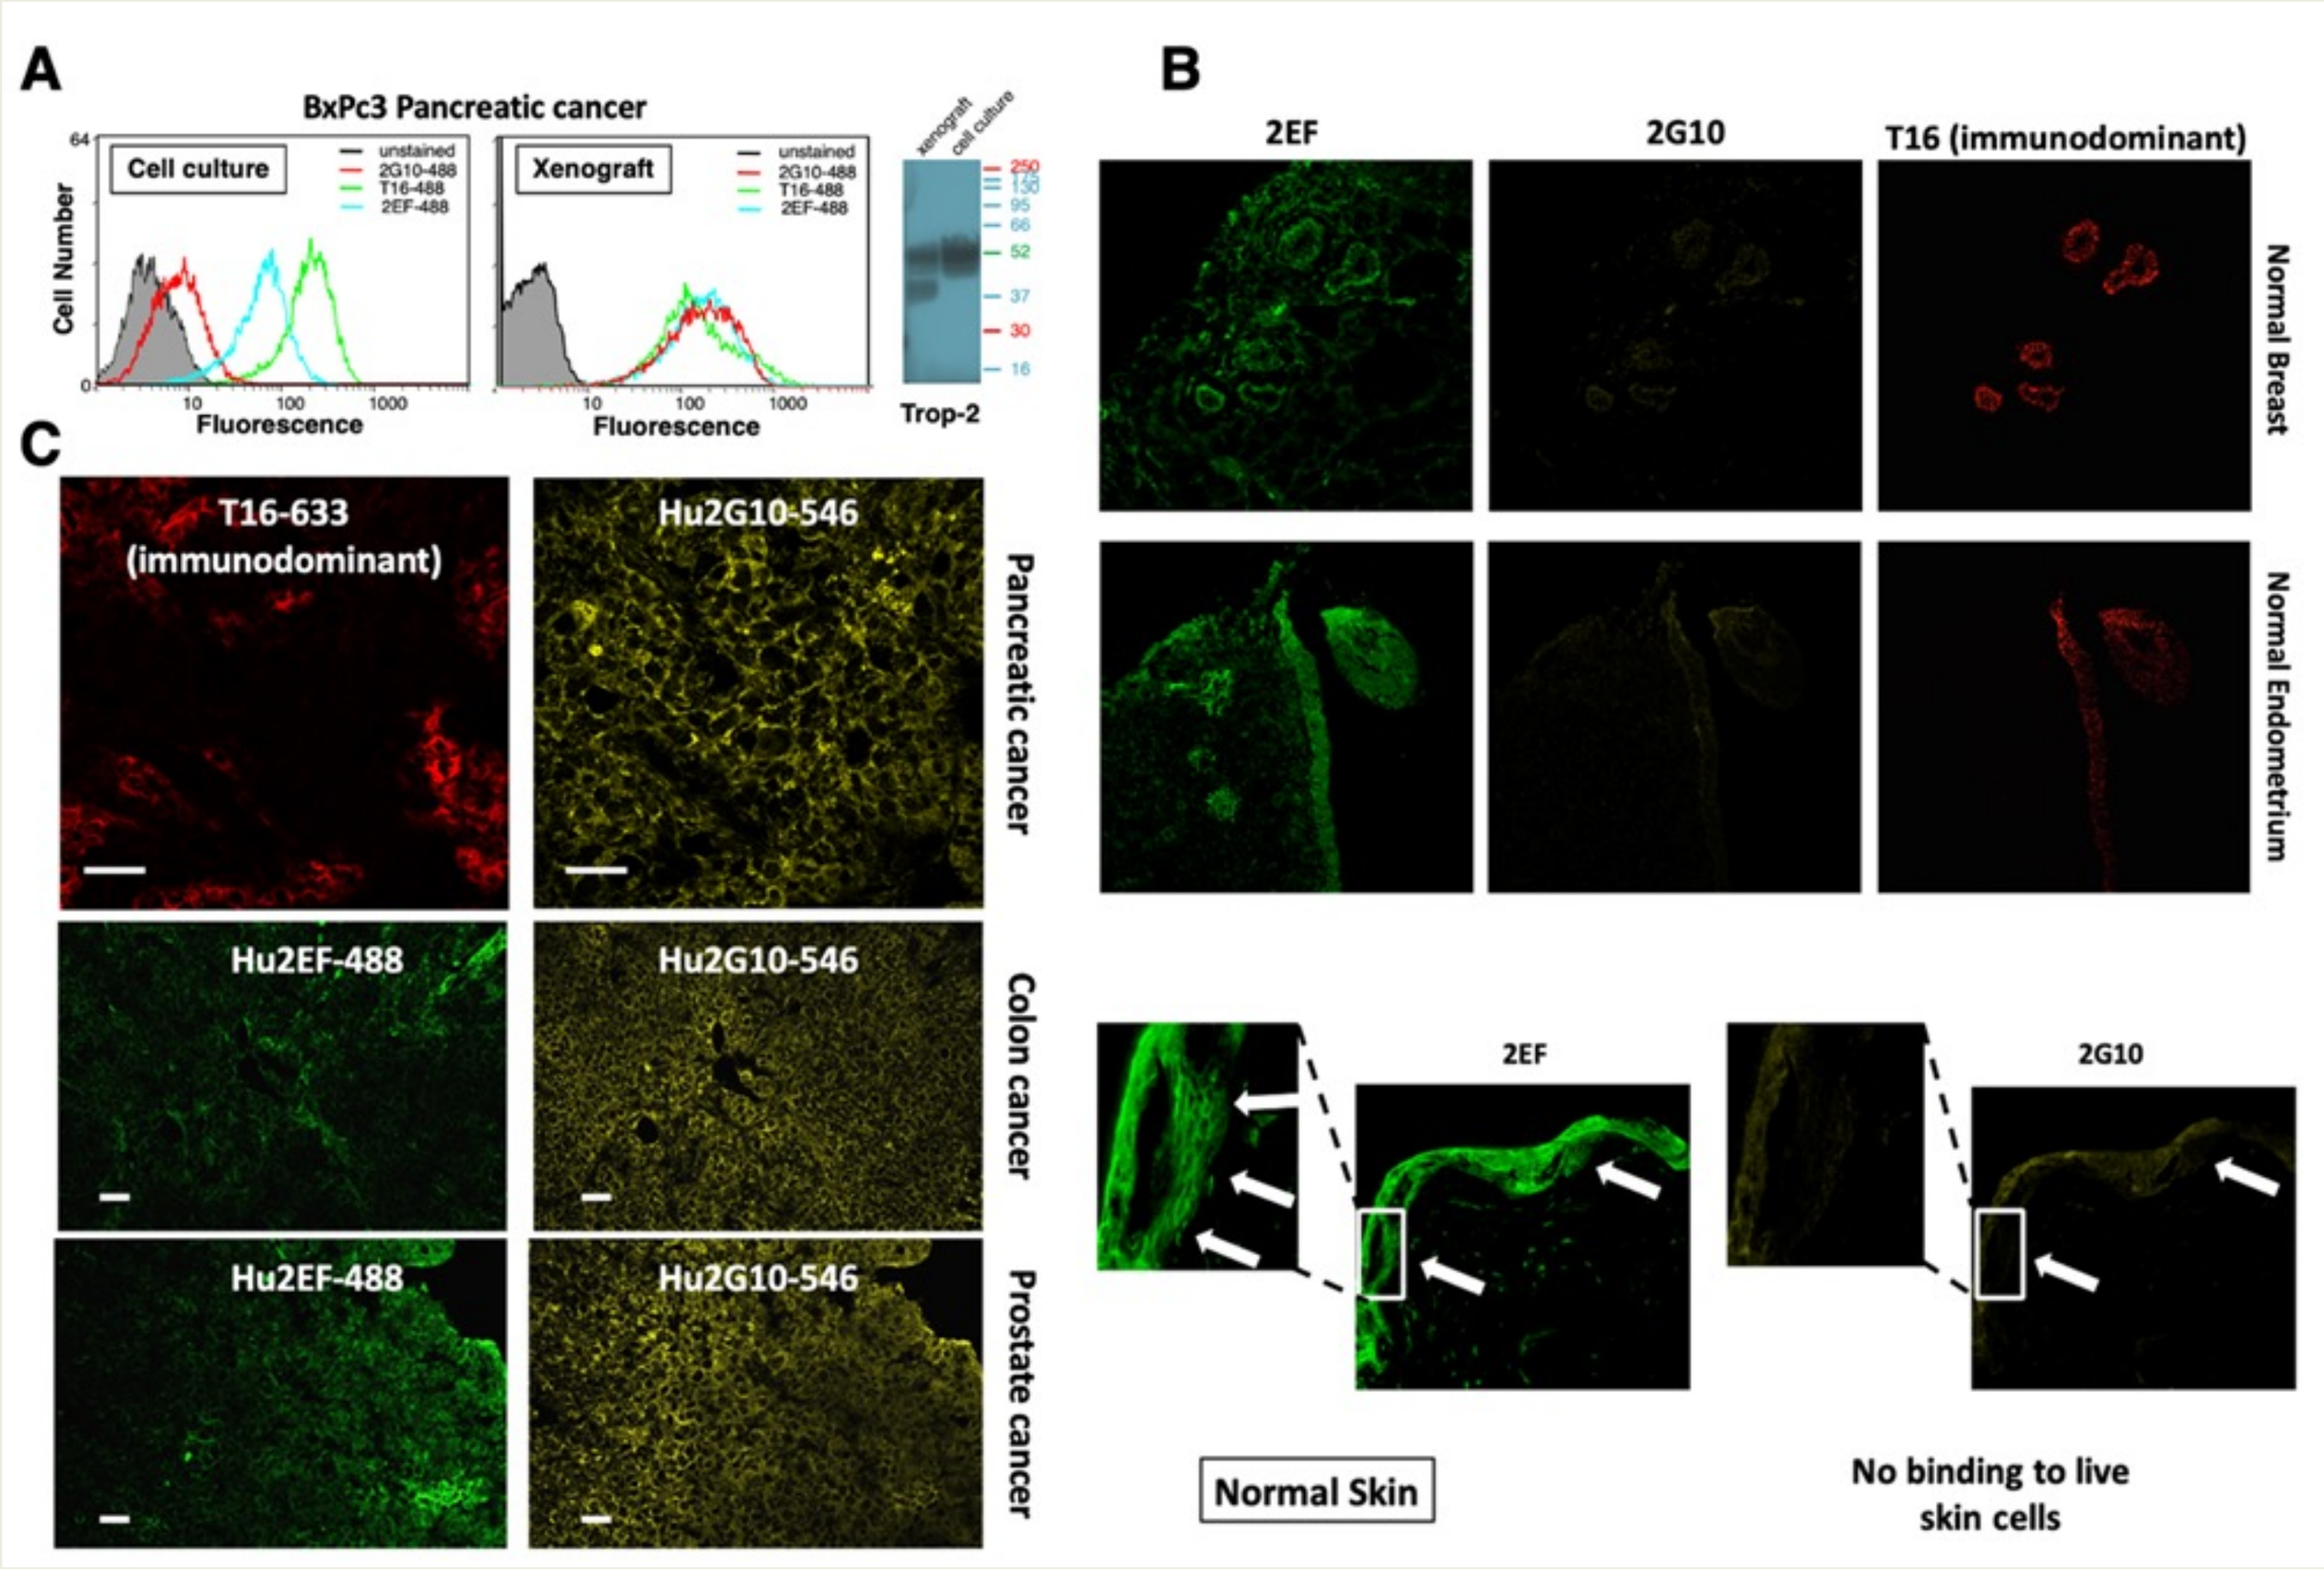
BxPc3 Pancreatic cancer
Cell culture
Xenograft
2EF
2G10
T16 (immunodominant)
Normal Breast
Normal Endometrium
T16-633 (immunodominant)
Hu2G10-546
Hu2EF-488
Pancreatic cancer
Colon cancer
Prostate cancer
Normal Skin
No binding to live skin cells

Figure 5. Recognition of cleaved/activated Trop-2 by Hu2G10 in cancer versus normal tissues. (A) (left, mid) Flow cytometry analysis of BxPC3 pancreatic cancer cells in vitro or ex-vivo. (right) Western blot analysis to assess the extent of Trop-2 cleavage. (B) Triple IF analysis of normal breast, endometrium, skin. The 2EF and the cancer-specific 2G10 signal is shown; white arrows indicate specific areas of Trop-2 expression, beneath the corneus layer. (C) Triple IF analysis of BxPc3 pancreatic, KM12SM colon, DU-145 prostate cancer xenografts.

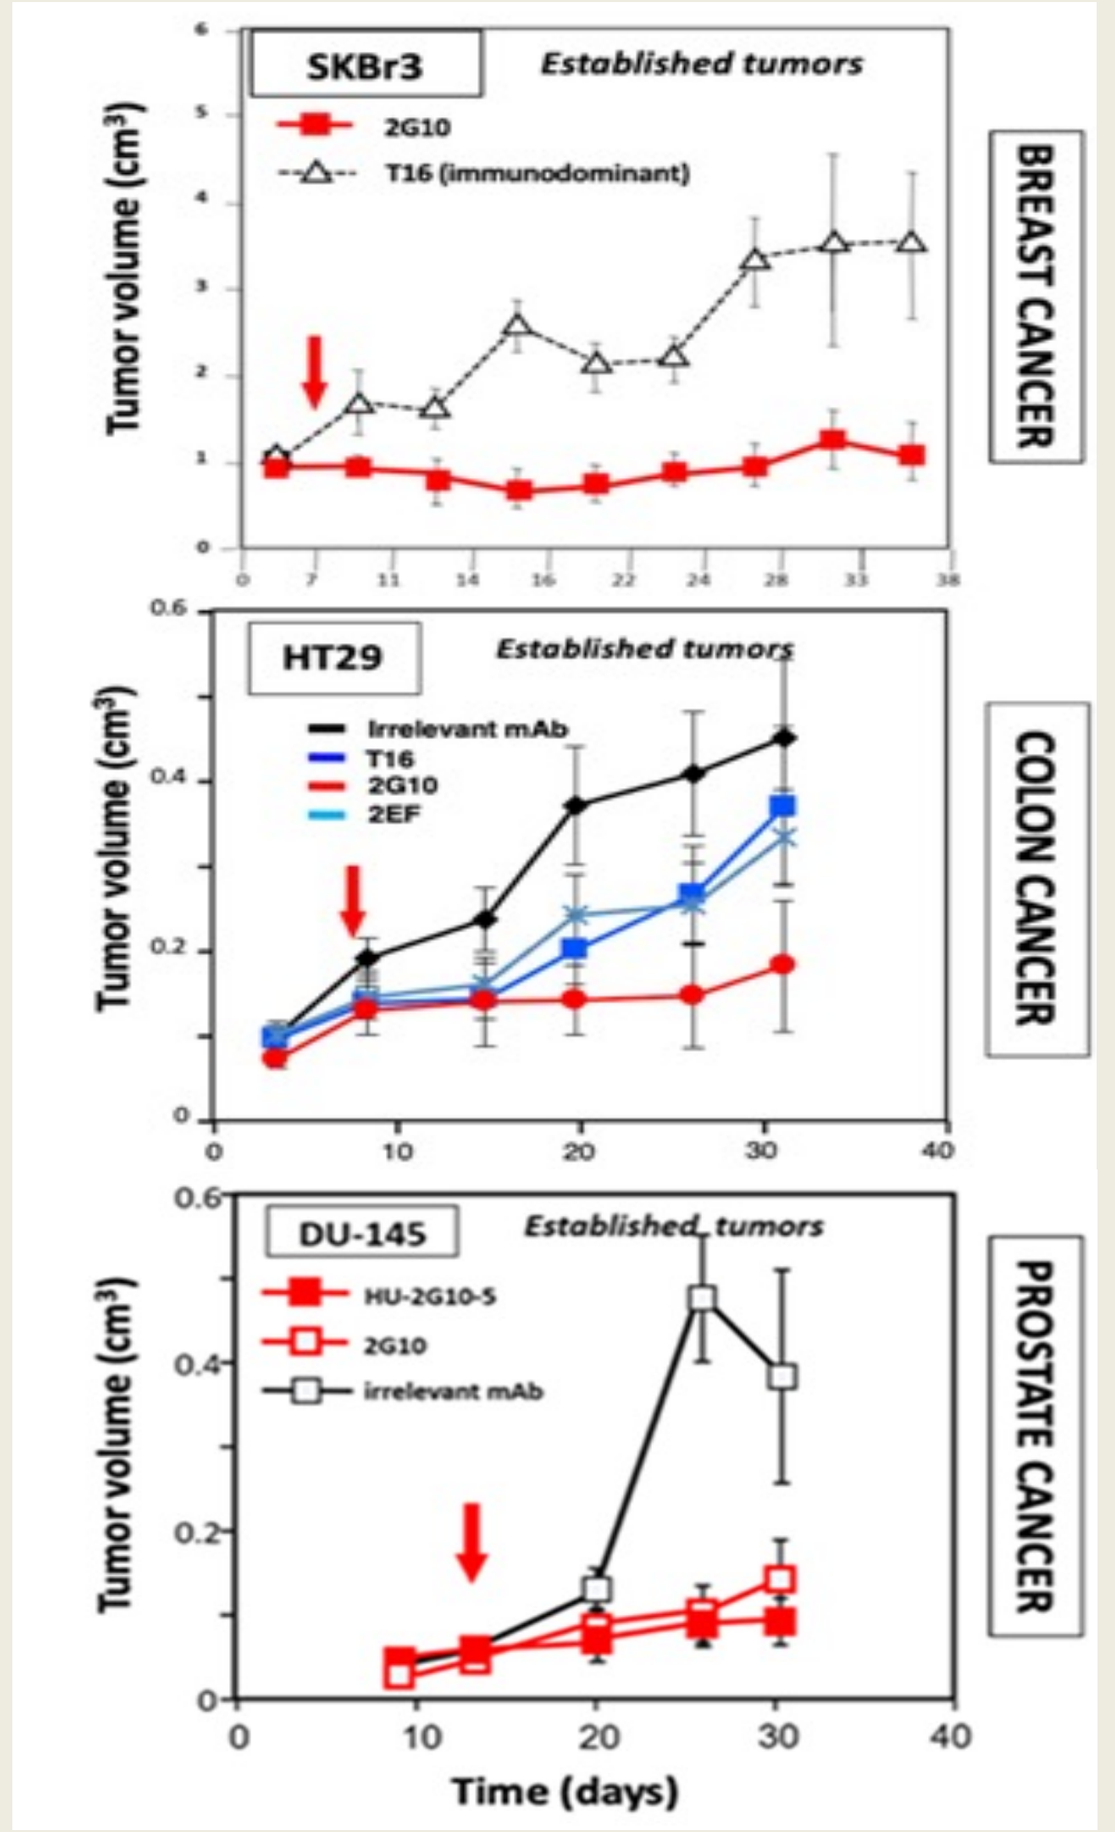
SKBr3
Established tumors
BREAST CANCER
HT29
COLON CANCER
DU-145
PROSTATE CANCER
Tumor volume (cm³)
Time (days)

Figure 6. 2G10 mAb immunotherapy of Trop-2-expressing cancer xenografts. Nude mice were injected sc with human cancer cells and weekly treated with 800 µg mAb (red arrow: first treatment).

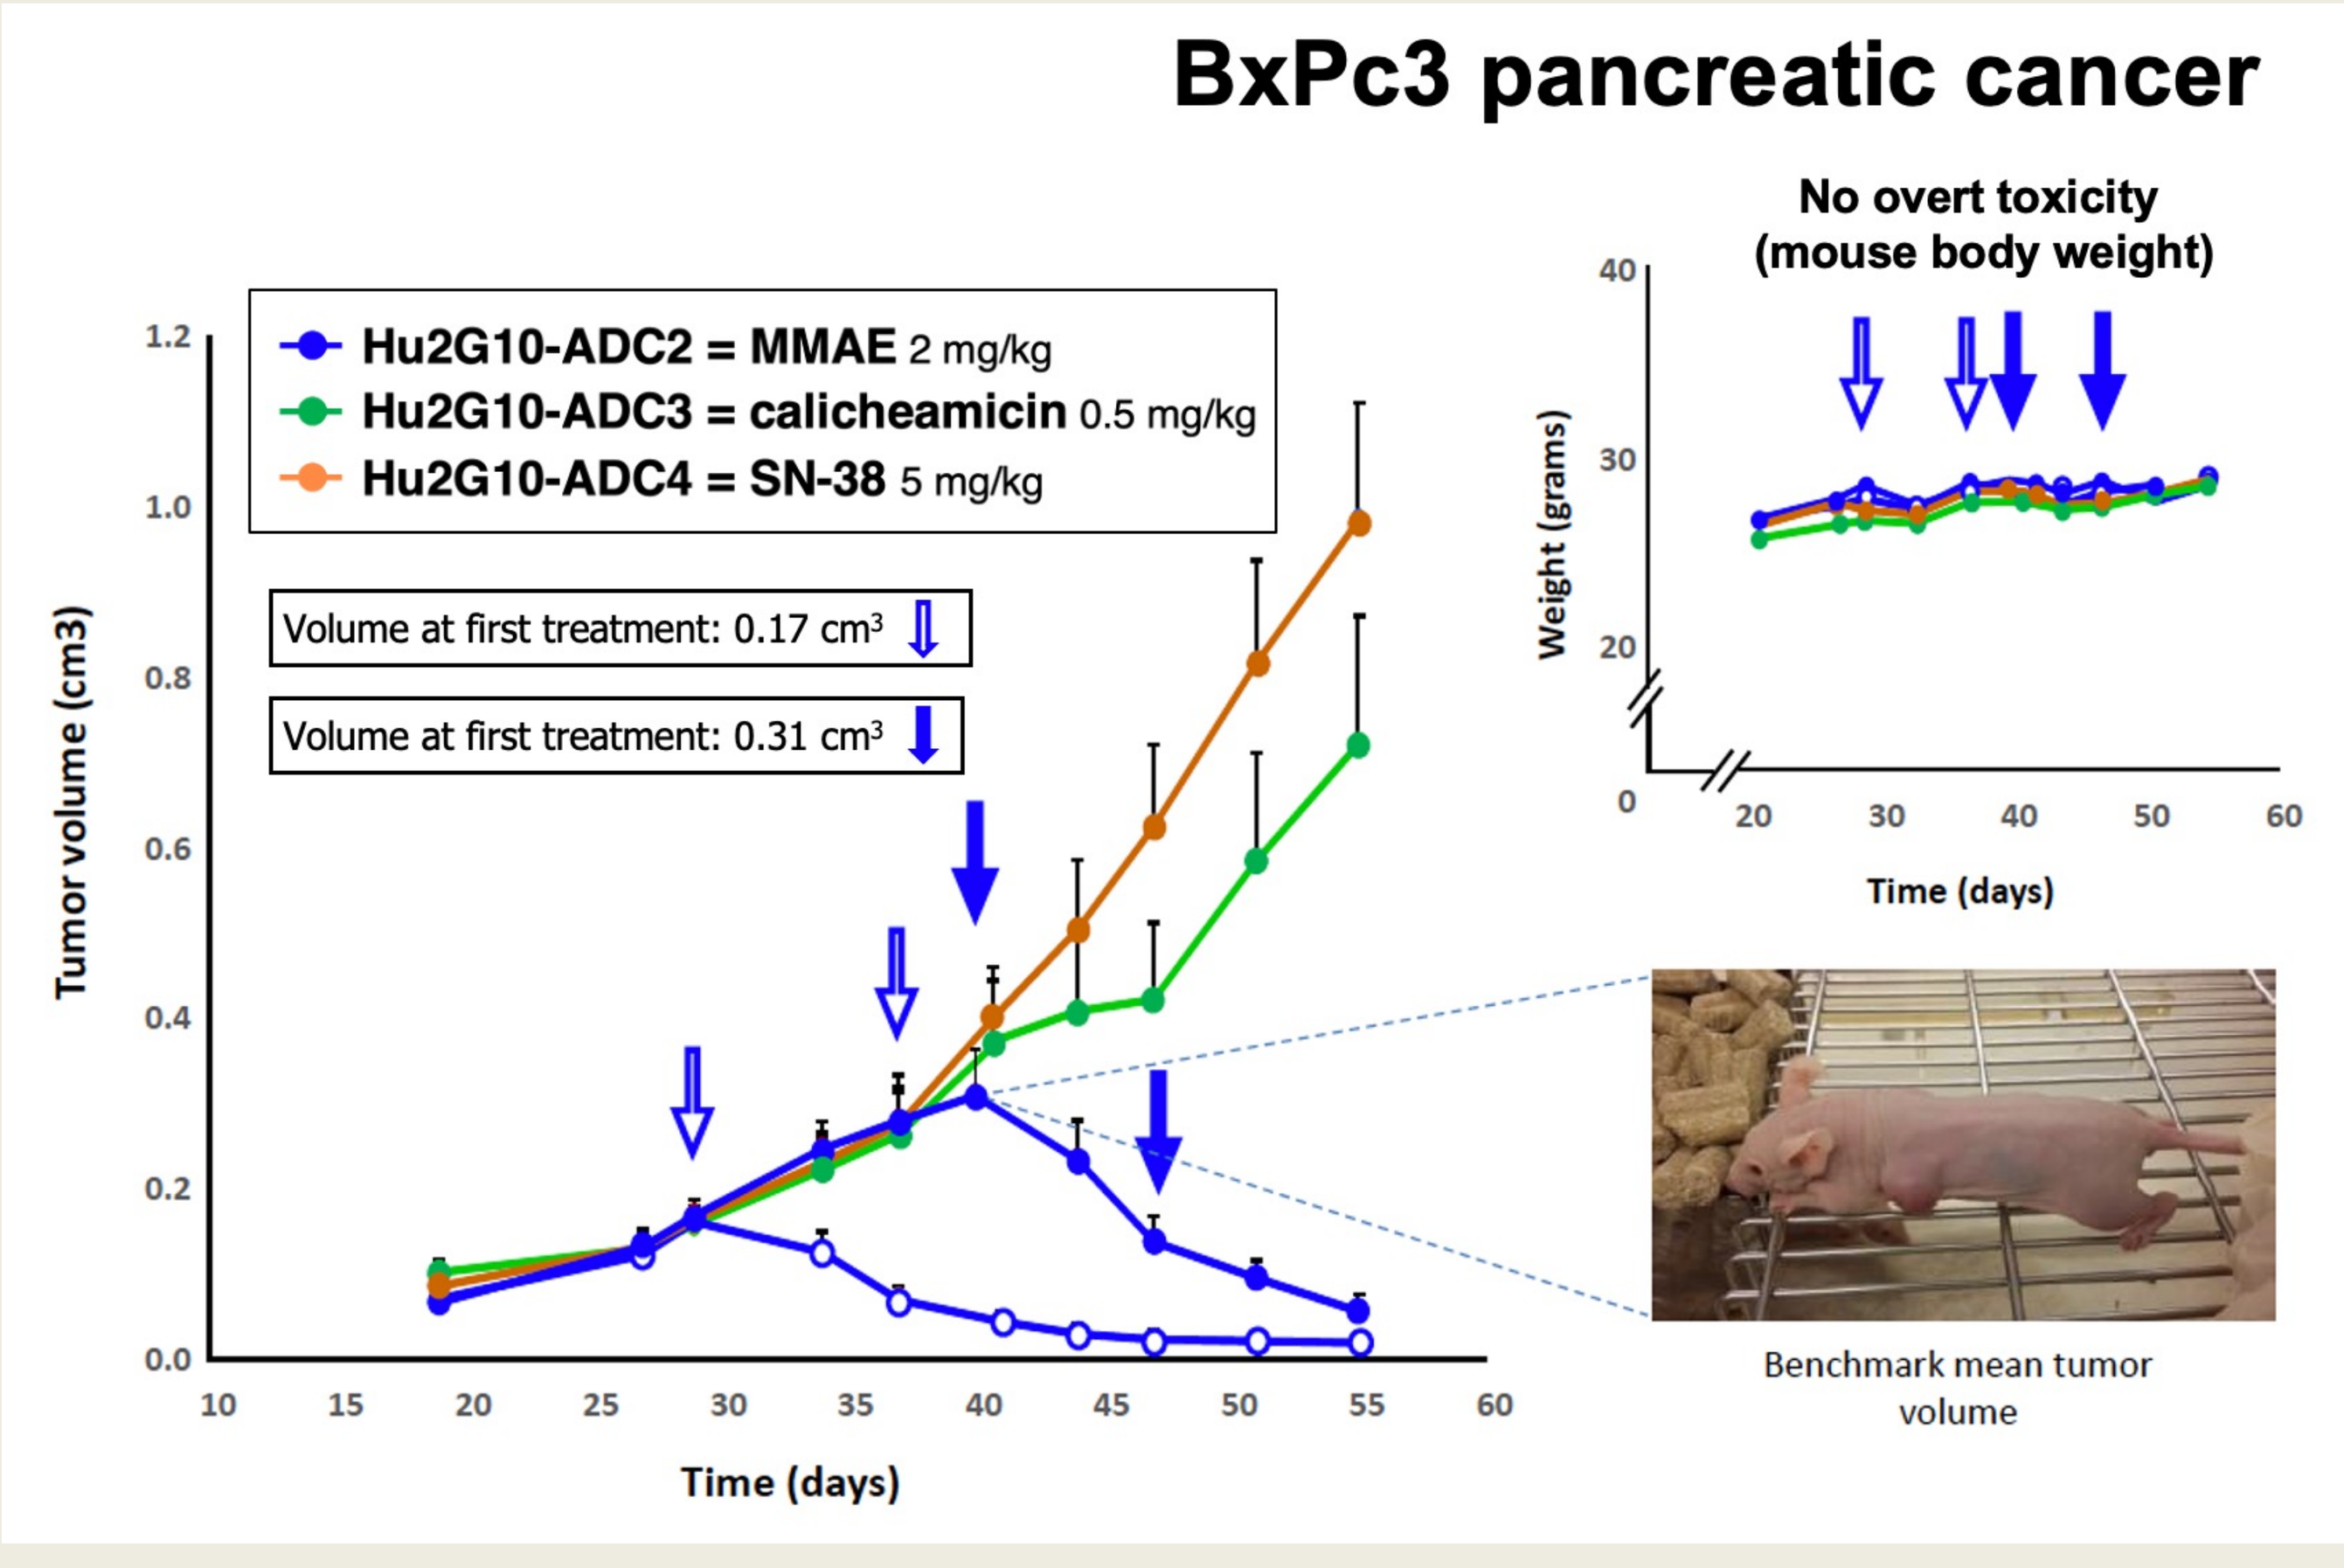
BxPc3 pancreatic cancer
Hu2G10-ADC2 = MMAE 2 mg/kg
Hu2G10-ADC3 = calicheamicin 0.5 mg/kg
Hu2G10-ADC4 = SN-38 5 mg/kg
Volume at first treatment: 0.17 cm³
Volume at first treatment: 0.31 cm³
No overt toxicity (mouse body weight)
Weight (grams)
Tumor volume (cm3)
Time (days)
Benchmark mean tumor volume

Figure 7. In vivo anticancer activity of Hu2G10 ADCs. Athymic mice subcutaneously injected with human cancer cells as indicated were randomized (n=16 per group) and treated as indicated (arrows). Mice in the control groups received an irrelevant isotype-matched mAb. Tumor volumes and body weights were plotted against time. The inset shows a representative 0.3 cm3 tumor. Error bars: ± SEM.

CONCLUSIONS

By targeting the activation site of Trop-2 we succeeded in generating the Hu2G10 Mab with extremely high affinity and potency while essentially devoid of toxicity. Our findings candidate Hu2G10 as a means for generating next-generation ADC, for enhanced selectivity and efficiency of payload delivery into tumor cells, while reducing the systemic toxicity caused by binding of normal cells. This is expected to lead to a paradigm shift for Trop-2-targeted therapy in patients.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com